Abstract # 3729

MP-470, a Rad51 Suppressor and Multiple TK Inhibitor, Retards Growth of Primary Human Glioblastoma Multiforma (GBM) Cells and Synergizes Effect of Temozolomide

Sarath Kanekal , Sabrina Lei, and Sanjeev Redkar
SuperGen Inc., Dublin CA, USA

Abstract

MP-470 suppresses Rad51, essential for repair of DNA double strand breaks, and multiple tyrosine kinases including mutant c-Kit, mutant PDGFR, mutant FLT3, and c-Met. In this study, primary GBM tumor specimens from eleven patients on standard of care treatment prior to surgery were used to evaluate MP-470's efficacy as a single agent and in combination with Temozolomide. The GBM cells were tested for cell viability and placed in soft agar 96-well plates in a humidified incubator (37°C, 5% CO2). MP-470 was evaluated at 1.25, 2.5, 5, 10 and 25 μM and Temozolomide at 100, 200, 400, 800 and 1600 μM, and MCF-7 cells were used as a control. After five days of incubation anchorage-independent growth was measured by Alamar Blue assay. Susceptibility to both drugs varied across the patients, MP-470's IC_{50} ranged from 0.05 to 29.6 μM (average: 4.6 μM) and Temozolomide IC_{50} ranged from 346.5 to 1958.8 μM (average = 1058 μM). While MP-470 was effective in all 11 tumor explants, Temozolomide demonstrated activity in 10 of the 11 explants. Furthermore, MP-470 was synergistic with Temozolomide in 8 of 11 explants, at two combinations (MP-470 2.5 μM + Temozolomide 800 M and MP-470 1.25 μM + Temozolomide 400 μM). In addition, exposure sequence tests showed synergy in 10 out of 11 explants when MP-470 (1.25 μM) was followed by Temozolomide (800 μM) 24+/-2h later. In conclusion, MP-470 inhibits growth of primary human GBM explants as a single agent and is synergistic with Temozolomide in combination; the combination is more effective when MP-470 is used first followed by Temozolomide.

Introduction

- MP-470 is a novel small molecule with potent activity against several tyrosine kinase targets: mutant c-Kit, mutant PDGFR, mutant Flt3 and c-Met.
- MP-470 also sensitizes cancer cells to DNA damaging agents and XRT by suppression of Rad51 and DNA double-strand break repair.
- Resistance of Glioblastoma Multiforme (GBM) to standard DNA damaging therapies is thought to be due to increased DNA repair and inhibited apoptosis.
- MP-470 was shown to sensitize GBM cell lines to XRT by inhibiting DNA double-strand break repair (Welsh et al.)
- MP-470 is currently being studied in a Phase I clinical trial and a Phase Ib five-arm combination trial with standard-of-care anti-cancer therapies in solid tumors.

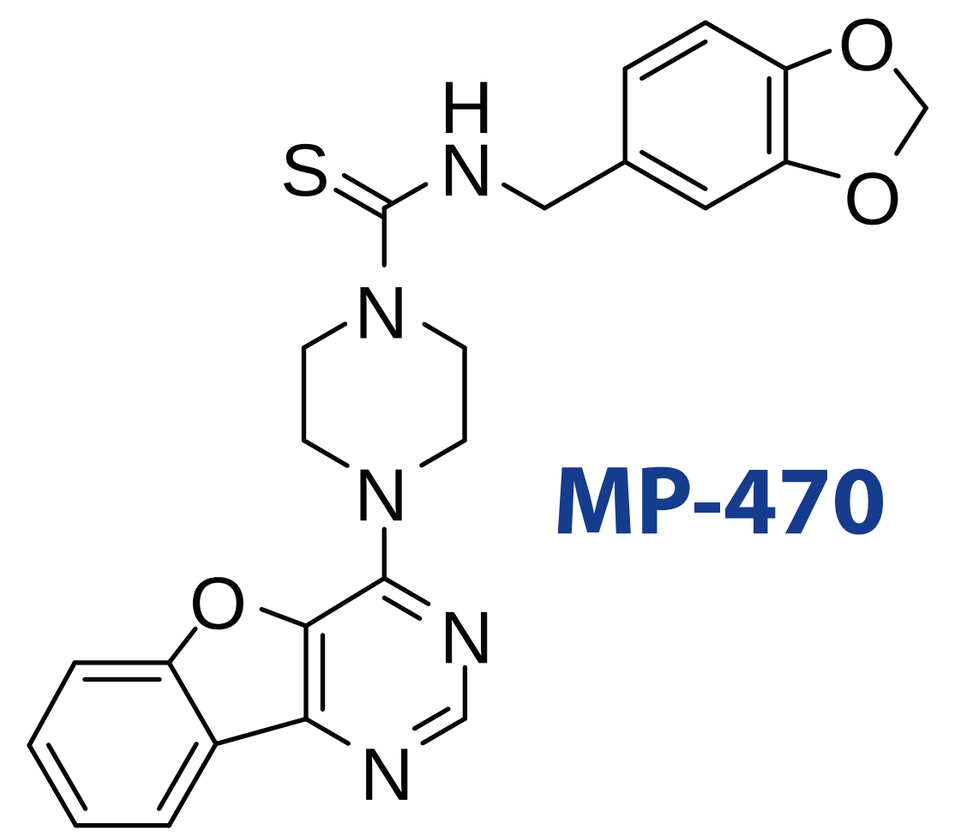

MP-470

Objective

In this study, primary GBM tumor specimens from eleven patients on standard of care treatment prior to surgery were used to evaluate MP-470's efficacy as a single agent and in combination with Temozolomide.

Materials & Methods

- Viably cryopreserved glioblastoma explants were used for this study.
- Tissues used in this study (Mosaic Laboratories, Lake Forest, CA) were collected from patients presenting for routine clinical evaluation in accordance with ethics review board-approved protocols at each clinical site.

Alamar Blue Assay (Mosaic Blue Assay™)

1. Viably cryopreserved glioblastoma explants in DMSO were thawed in a manner to maintain viability.
2. Tumor cells were evaluated for viability and percent tumor prior to plating in soft agar in a 96-well plate in the presence of MP-470 (range 25-0.0390625 μM) or temozolomide (range 1600- 12.5 μM).
3. Anchorage-independent growth was measured by fluorescence using Alamar Blue (Invitrogen) on day 5. Percent inhibition was calculated by comparing the average corrected MP-470 and temozolomide results to the average corrected no drug treatment results.

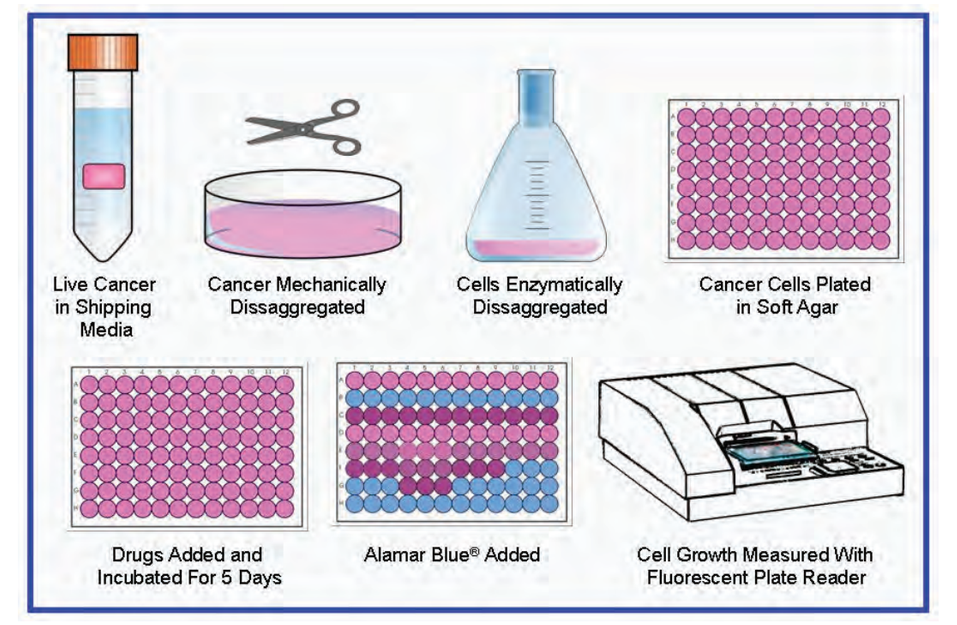
Live Cancer in Shipping Media
Cancer Mechanically Disaggregated
Cells Enzymatically Disaggregated
Cancer Cells Plated in Soft Agar
Drugs Added and Incubated For 5 Days
Alamar Blue® Added
Cell Growth Measured With Fluorescent Plate Reader

Combination Index

The combination index (CI) was calculated using CalcuSyn (V2.0, Biosoft, Cambridge, United Kingdom) using the median-effect analysis derived by Chou and Talalay and the following equation:

$$CI = \frac{D^1}{D^1_x} + \frac{D^2}{D^2_x} + \frac{D^1.D^2}{D^1_x.D^2_x}$$

where D1 and D2 are the doses of drug 1 and drug 2 that have x% effect when used in combination, and Dx^1 and Dx^2 are the doses of drug 1 and drug 2 that have the same x% effect when used alone
Synergy, additivity, and antagonism are defined as
CI < 1 – Synergy
CI = 1 – Additivity
CI > 1 - Antagonism

Clinical Information on Tumors

Mosaic ID	Tumor	Site	Age	Gender	Race	Prior Therapy	Comments
Titration Phase							
ML0608035	Glioblastoma	Occipital Lobe	51	F	White	Unknown	
ML0703081	Glioblastoma	Brain	44	M	White	Yes	Dexamethasone
ML0710088	Glioblastoma	Brain	59	F	White	No	
Testing Phase							
ML0610017	Glioblastoma	Brain	59	M	White	Unknown	
ML0612059	Glioblastoma	Brain	62	F	White	Unknown	
ML0701091	Glioblastoma	Brain	66	M	White	Unknown	
ML0703049	Glioblastoma	Brain	NA	F	White	Yes	Dexamethasone
ML0703063	Glioblastoma	Brain	48	M	White	Yes	Dexamethasone
ML0704021	Glioblastoma	Brain	48	F	White	Yes	Dexamethasone
ML0705060	Glioblastoma	Brain	60	M	White	Unknown	
ML0711030	Glioblastoma	Brain	35	M	White	Yes	Dexamethasone
ML0711050	Glioblastoma	Brain	43	F	White	Yes	Dexamethasone
ML0711165	Glioblastoma	Left Temporal Lobe	61	M	White	No	
ML0801114	Glioblastoma	Brain	43	F	Unknown	Yes	Dexamethasone

Results

Titration of MP-470 and Temozolomide in GBM Primary Cells

MP-470

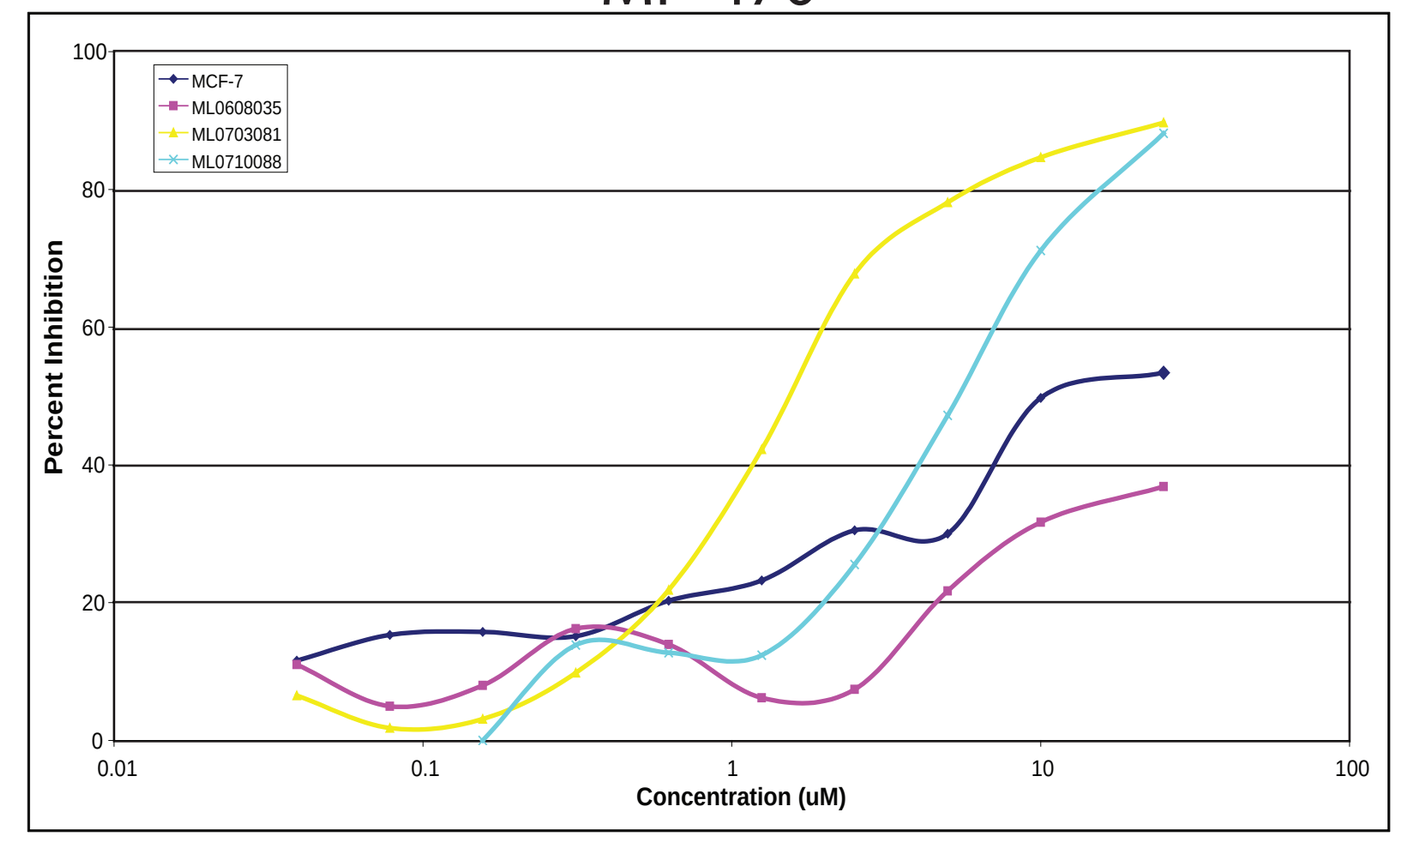
MCF-7
ML0608035
ML0703081
ML0710088
Percent Inhibition
Concentration (uM)
0.01
0.1
1
10
100

Temozolomide

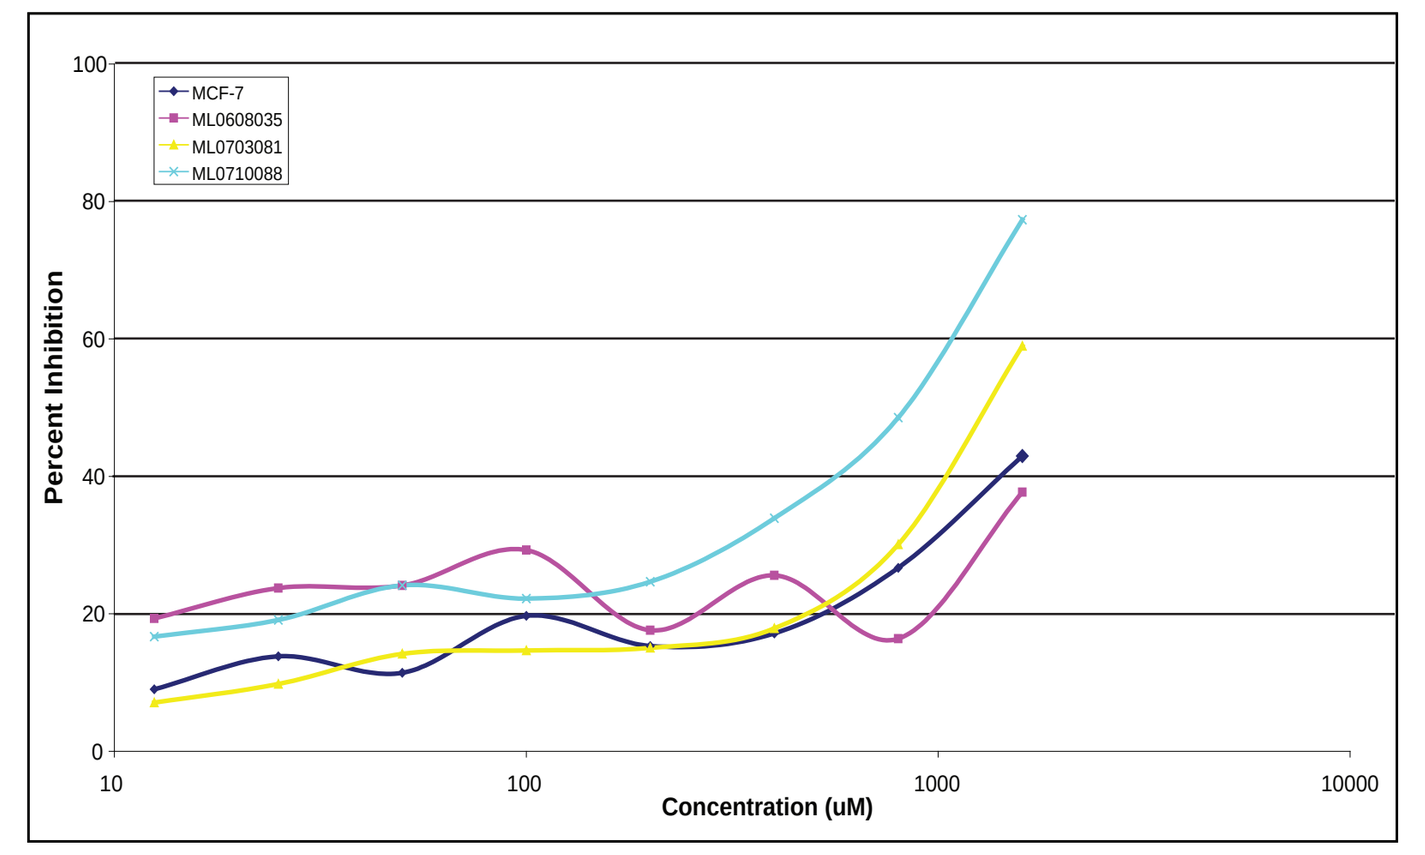
MCF-7
ML0608035
ML0703081
ML0710088
Percent Inhibition
Concentration (uM)
10
100
1000
10000

Outline of single agent and combination drug concentrations

Outline of Drug Concentrations	MP470 Concentrations	Temozolomide Concentrations
MP-470 Single Agent	25, 10, 5, 2.5, and 1.25 μM	NA
Temozolomide Single Agent	NA	1600, 800, 400, 200, and 100 μM
MP-470 + Temozolomide Combination	5, 2.5, and 1.25 μM	800, 400, and 200 μM
MP-470 (Day 0); Temozolomide (Day 1)	2.5 μM	1600, 800, 400, 200, and 100 μM
MP-470 (Day 0); Temozolomide (Day 1)	25, 10, 5, 2.5, and 1.25 μM	800 μM

Single agent IC_{50} of MP-470 and Temozolomide in 11 GBM Primary Samples

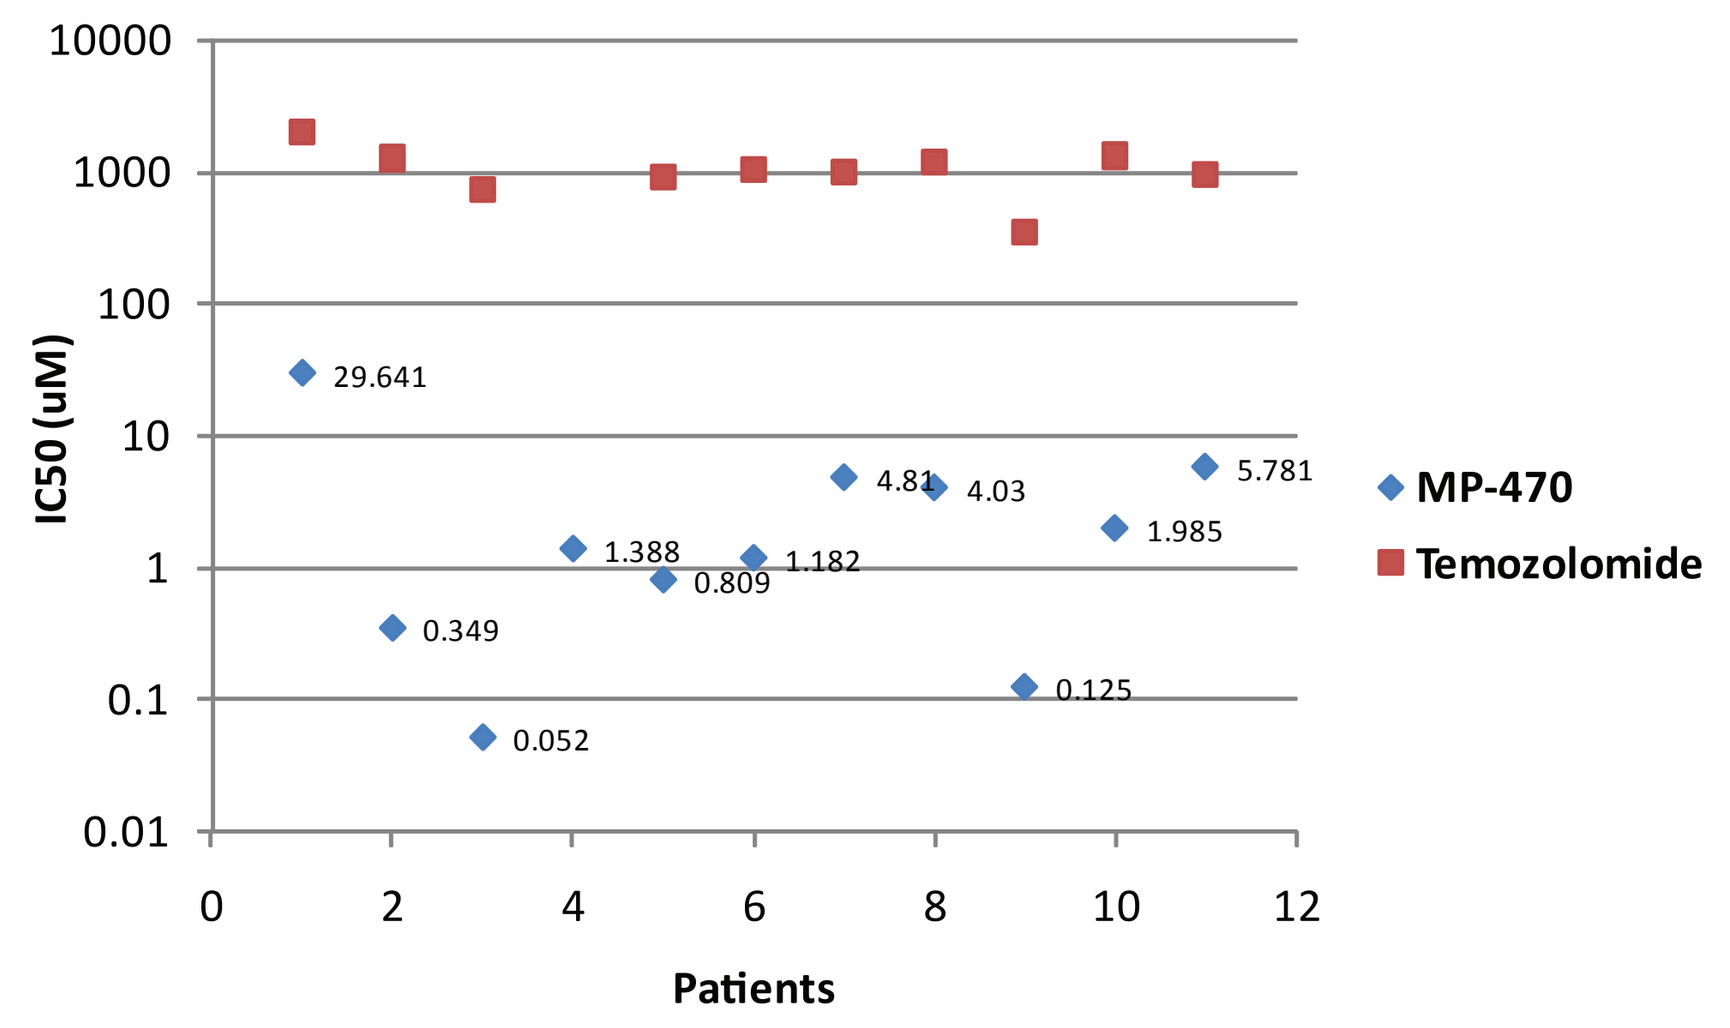
IC50 (uM)
Patients
29.641
0.349
0.052
1.388
0.809
1.182
4.81
4.03
0.125
1.985
5.781
MP-470
Temozolomide

Average effect of MP-470 and Temozolomide on 11 Glioblastoma Explants in the Almar Blue Assay.

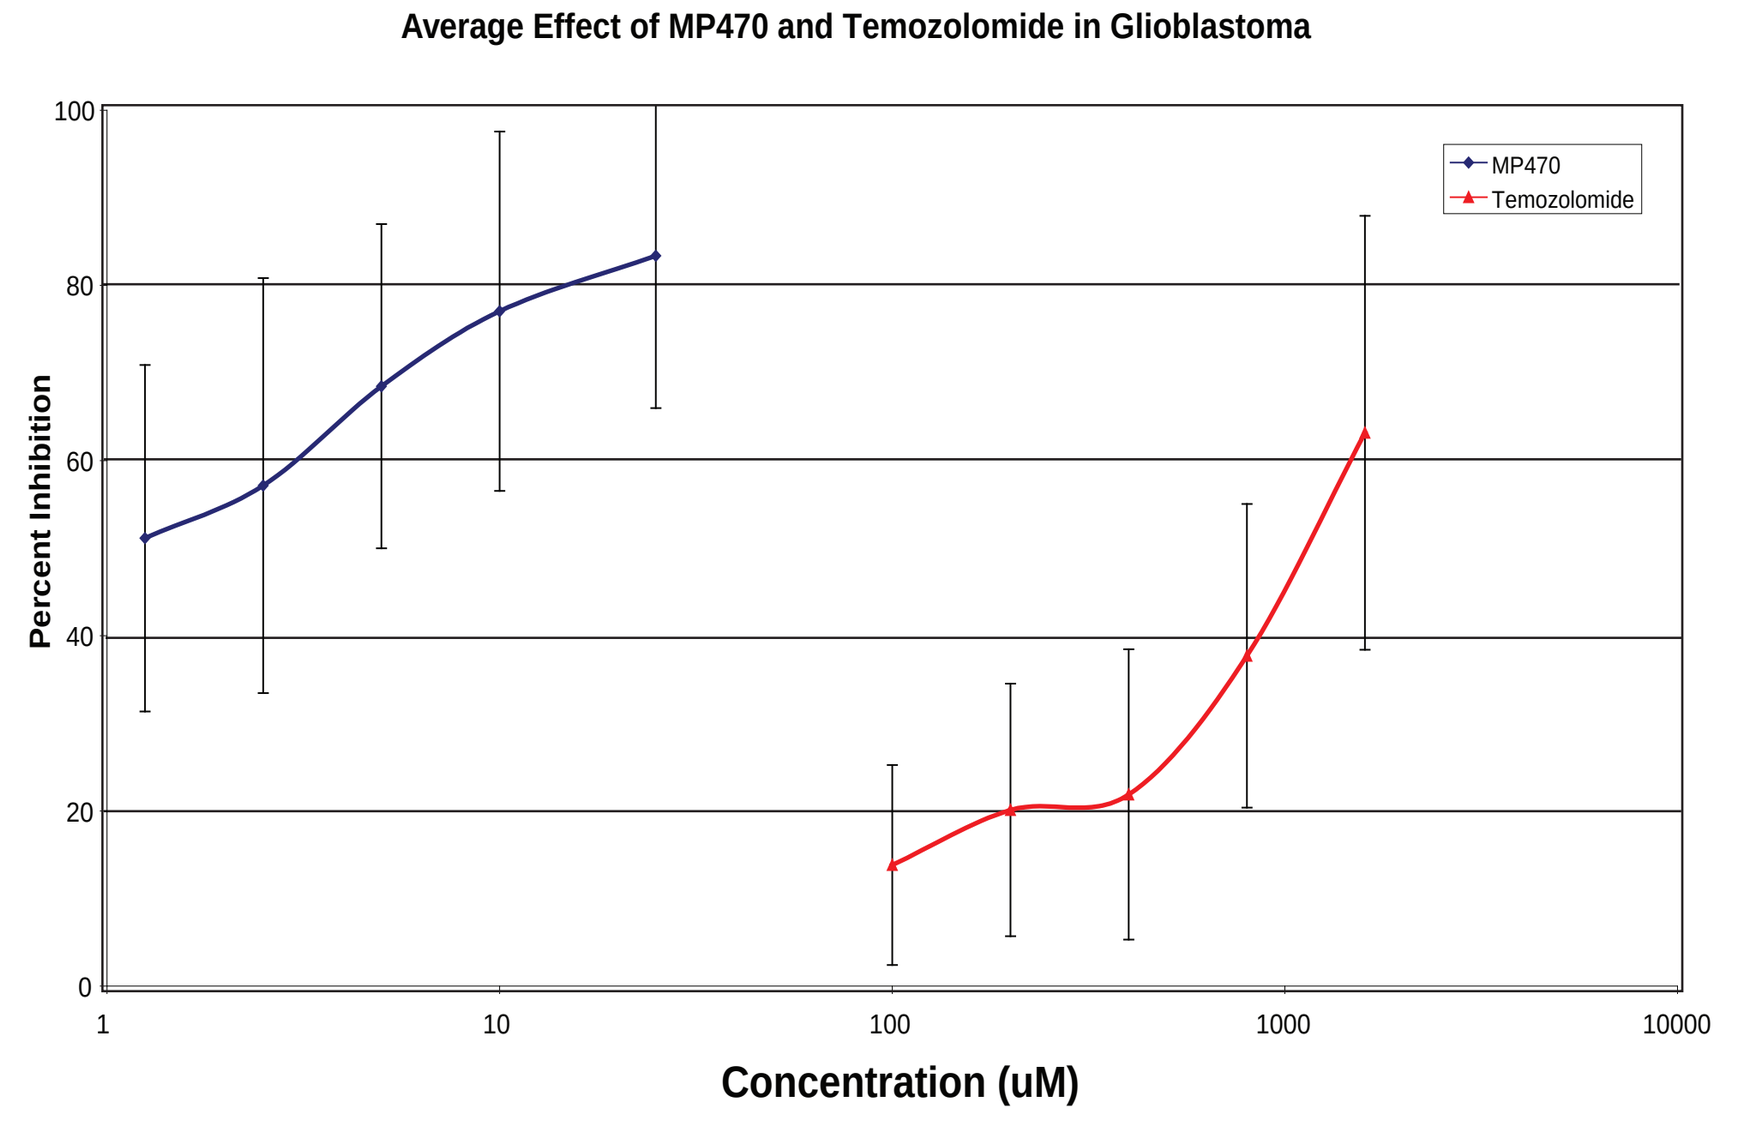
Average Effect of MP470 and Temozolomide in Glioblastoma
MP470
Temozolomide
Percent Inhibition
Concentration (uM)

Median Normalized CI Values

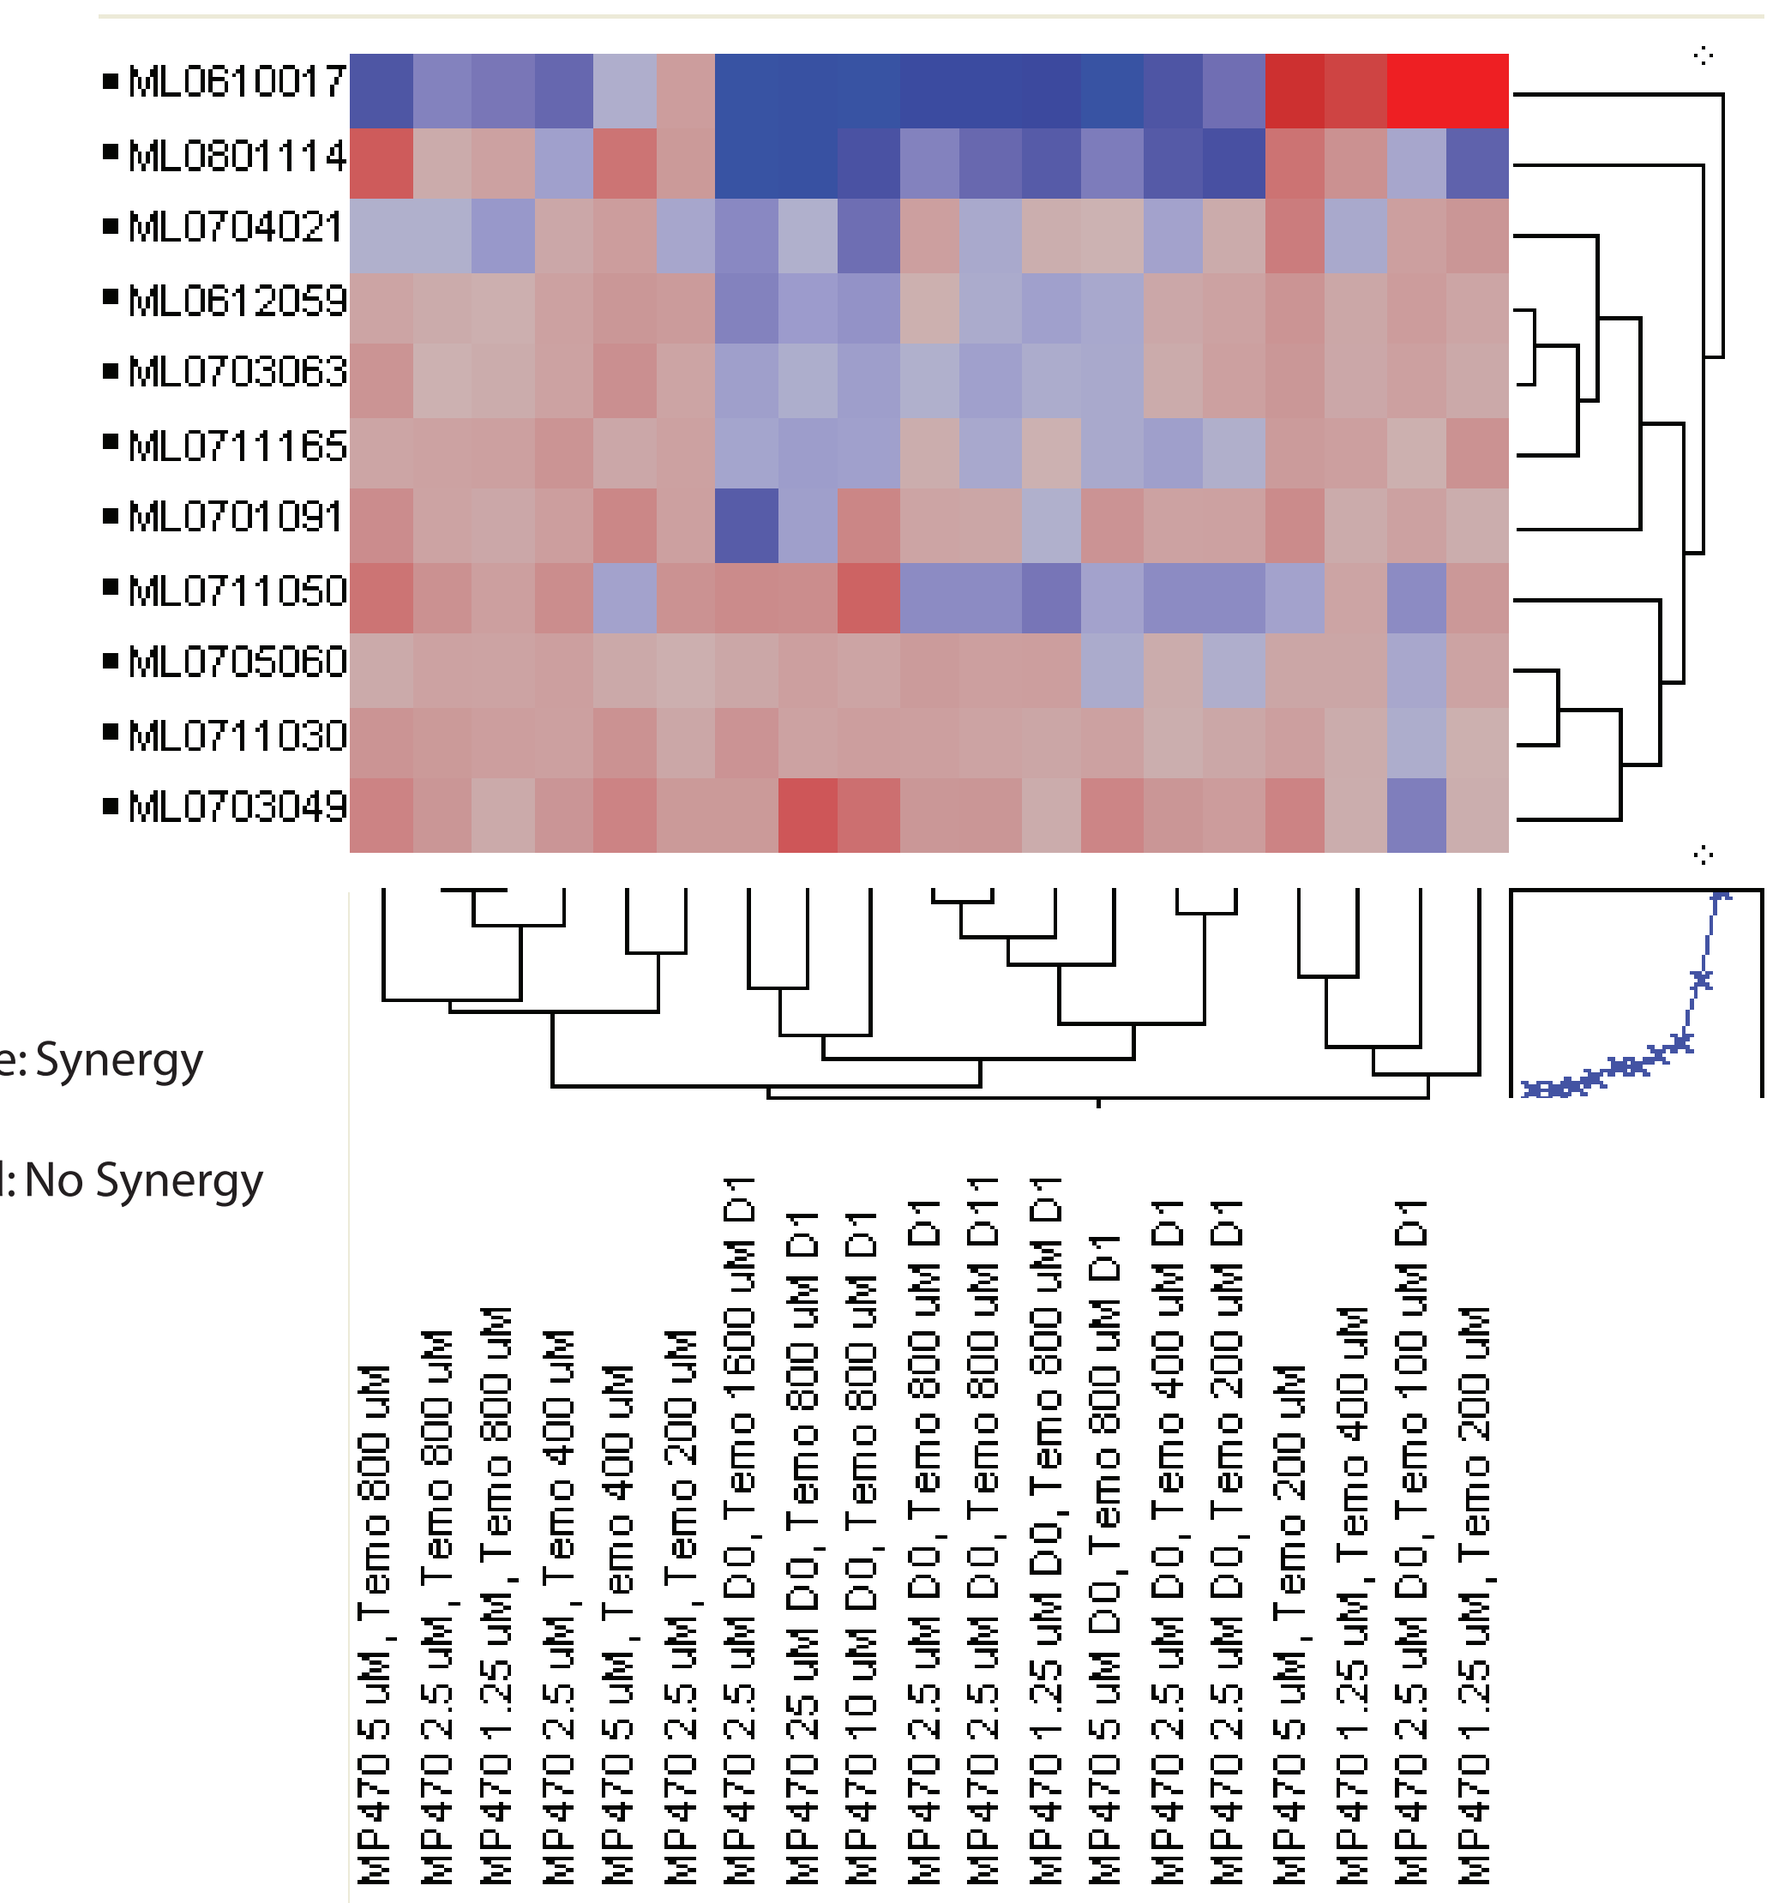
ML0610017
ML0801114
ML0704021
ML0612059
ML0703063
ML0711165
ML0701091
ML0711050
ML0705060
ML0711030
ML0703049
MP470 5 uM , Temo 800 uM
MP470 2.5 uM , Temo 800 uM
MP470 1.25 uM , Temo 800 uM
MP470 2.5 uM , Temo 400 uM
MP470 5 uM , Temo 400 uM
MP470 2.5 uM , Temo 200 uM
MP470 2.5 uM D0 , Temo 1600 uM D1
MP470 25 uM D0 , Temo 800 uM D1
MP470 10 uM D0 , Temo 800 uM D1
MP470 2.5 uM D0 , Temo 800 uM D1
MP470 2.5 uM D0 , Temo 800 uM D11
MP470 1.25 uM D0 , Temo 800 uM D1
MP470 5 uM D0 , Temo 800 uM D1
MP470 2.5 uM D0 , Temo 400 uM D1
MP470 2.5 uM D0 , Temo 200 uM D1
MP470 5 uM , Temo 200 uM
MP470 1.25 uM , Temo 400 uM
MP470 2.5 uM D0 , Temo 100 uM D1
MP470 1.25 uM , Temo 200 uM

Grey to Blue: Synergy
Grey to Red: No Synergy

Conclusions

- MP-470 demonstrated activity in all 11 glioblastoma explants tested.
 - IC_{50} values for MP470 ranged from 0.052 μM to 29.641 μM (average = 4.56 μM).
- Temozolomide demonstrated activity in 10 out of 11 glioblastoma explants tested.
 - One specimen treated with temozolomide (ML0703049) demonstrated complete resistance to temozolomide at all 5 concentrations tested.
 - IC_{50} values for temozolomide ranged from 346.479 μM to 1958.750 μM (average = 1058.19 μM).
- Low μM MP-470 was synergistic when combined simultaneously or sequentially with Temozolomide in 10 out of 11 explants.
- A greater degree of synergy was observed when MP-470 and Temozolomide were tested sequentially compared to simultaneously.

References

1. J. Welsh, D. Mahadevan, D. Bearss, B. Stea. Sensitization of a Glioblastoma Multiforme (GBM) cell Line by MP470: A novel c-Met antagonist. International Journal of Radiation Oncology/Biology/Physics, Vol 69, Issue 3, Pages S100-S100
2. Friedman HS, Kerby T, Calvert H. Temozolomide and treatment of malignant glioma. Clin Cancer Res. 6:2585–97, 2000.
3. A. W. Tolcher et al. Clinical responses in highly refractory solid tumor patients with oral MP-470, a multi-targeted tyrosine kinase inhibitor, in combination with standard of care chemotherapy regimens: preliminary report from a multi-institutional Phase-Ib clinical trial. Proceedings from the EORTC-NCI-AACR Conference 2008.
4. R. Tibes et al. A Phase-1 study of MP-470, a novel orally bioavailable small molecule with Rad51 suppression activity. Proceedings from ESMO Annual Meeting 2008.
5. A. W. Tolcher et al. MP470, a Potent Oral Rad51 Suppressor Is Safe and Tolerable in First-in-Human Study. Proceedings from AACR Annual Meeting 2008.

Acknowledgements

Dr. Christopher Kerfoot and Lisa Dauffenbach at Mosaic Laboratories, Lake Forest, CA
Jeremy Lamb, Medical Writer, SuperGen, Inc.
Dr. Pietro Taverna, Associate Director, Translational Pharmacology, SuperGen, Inc.
Dr. Stuart Hwang, Director, Business Development, SuperGen, Inc.

AACR 100th Annual Meeting, 2009

SuperGen®
MP470PB9(905)